Evaluation of Vilobelimab as a Cost-Effective Option to Treat Severely Ill Mechanically Ventilated Patients with COVID-19

Daniel C. Malone, PhD, FAMCP[1]; Roger Seheult, MD[2]; Diana Brixner, RPh, PhD[1]; Gary Oderda, PharmD, MPH[1]; Bruce P. Burnett[3]; Joseph Biskupiak, PhD, MBA[1]
[1]University of Utah College of Pharmacy, SLC UT[2]; University of California Riverside School of Medicine, Riverside CA[3]; InflaRx Pharmaceuticals Inc., Ann Arbor MI

BACKGROUND

- Vilobelimab (Gohibic®), an anti-C5a-specific complement factor blocker, has an FDA Emergency Use Authorization (EUA) for the treatment of COVID-19 in hospitalized adults when initiated within 48 hours of receiving invasive mechanical ventilation (IMV) or extracorporeal membrane oxygenation (ECM0).[1]
- The PANAMO Phase 3 global study evaluated vilobelimab in critically ill adult COVID-19 patients (N=368), and included the use of corticosteroids (99%), anti-thrombotic agents (98%), and immunomodulators (IMs) (19%; tocilizumab or baricitinib).
- Vilobelimab significantly reduced 28-day all-cause mortality (32%) compared to placebo (42%) when used in addition to standard-of-care (SoC).[2] (HR 0.67, 95%CI:0.48-0.96, p=0.027)
- In a post-hoc analysis, vilobelimab administered with prior or concomitant use of IMs was shown to potentially provide additional survival benefit on top of vilobelimab+SoC (n=34) vs placebo+SoC (n=37).[3]
- 28-day all-cause mortality: 6.3% (vilobelimab+SoC) vs 40.9% (placebo+SoC) (HR 0.13; 95%CI:0.3-0.56, p=0.006)

OBJECTIVE

- Evaluate the cost-effectiveness (CE) of vilobelimab added to standard of care based on the PANAMO Phase 3 clinical trial data in both the total study population and in the subpopulation receiving any prior or concomitant treatment with the immunomodulators tocilizumab or baricitinib.

METHODS

- A cost-effectiveness model was developed to compare vilobelimab+SoC to SoC alone for hospitalized severe COVID-19 patients requiring IMV or ECMO.
- A lifetime model used a modified societal perspective.
 - Included a short-term acute care decision tree followed by a post-discharge two-state Markov cohort model with a cycle length of 1 month and half-cycle correction (see Figure 1).
- Model estimated progression from severe COVID-19 to survival or death and the receipt of renal replacement therapy.
- Survivors then transitioned to the Markov model where transition probability from alive to death was based on CDC life-tables.[4]

Figure 1: Model structure

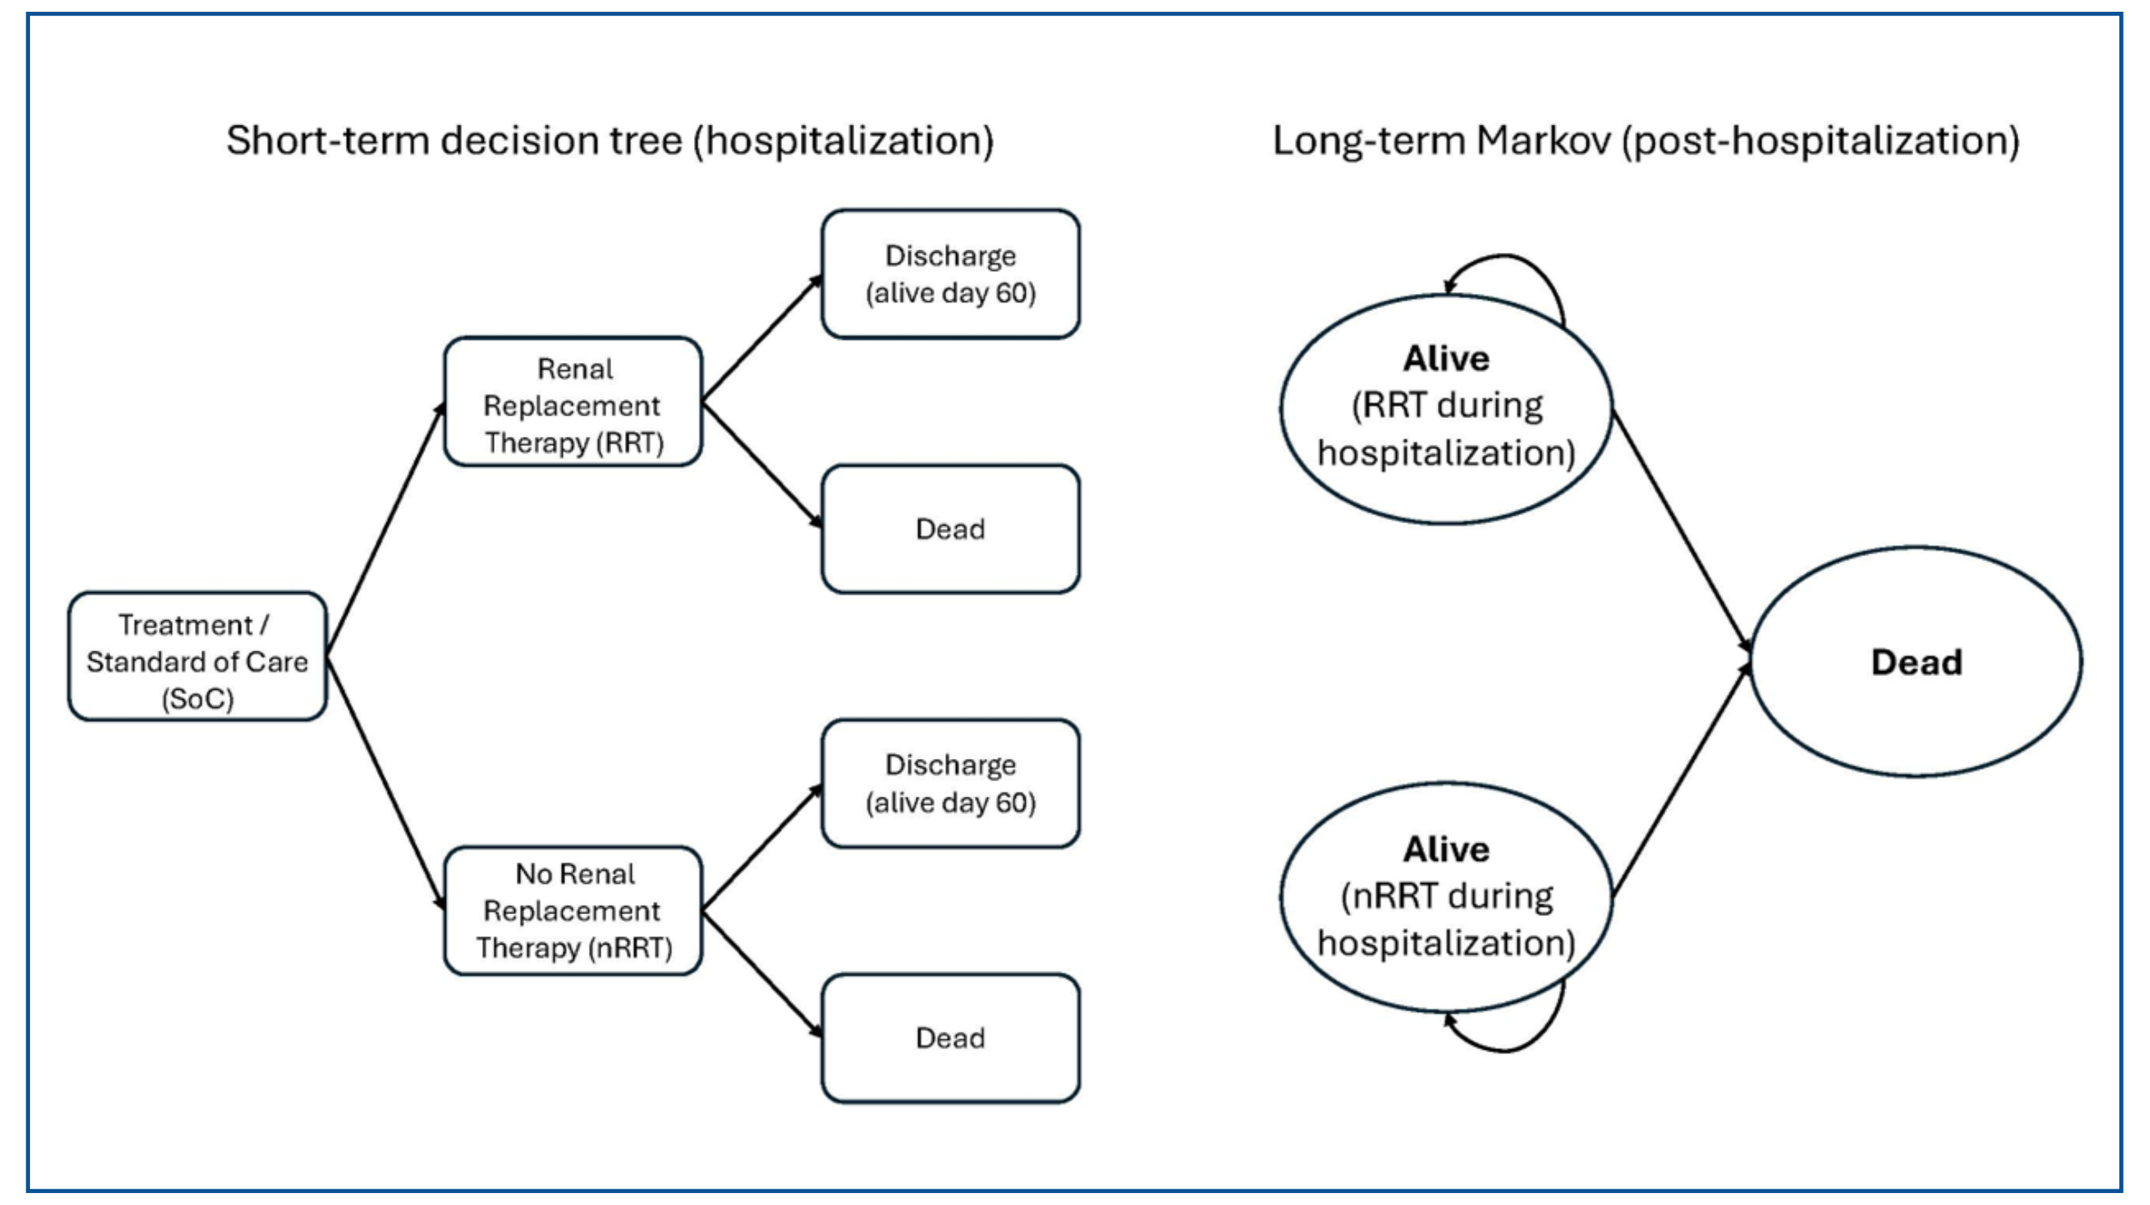
Short-term decision tree (hospitalization)
Long-term Markov (post-hospitalization)
Treatment / Standard of Care (SoC)
Renal Replacement Therapy (RRT)
No Renal Replacement Therapy (nRRT)
Discharge (alive day 60)
Dead
Discharge (alive day 60)
Dead
Alive (RRT during hospitalization)
Alive (nRRT during hospitalization)
Dead

- Model inputs:
 - Clinical outcomes were derived from the PANAMO Phase 3 study:
 - 60-day all-cause mortality data
 - Renal replacement therapy (RRT) at Day 28
 - Cost of infusion was not considered in the analysis because all patients were hospitalized, receiving supportive care
 - Tables 1, 2, and 3 contain model inputs:
 - Sheinson – CE framework for hospitalized COVID-19 patients[5]
 - Putri – premature mortality cost[6]

This study was funded by a research grant from InflaRx Pharmaceuticals, Inc. to Millcreek Outcomes Group, LLC.

AMCP Nexus 2024, Las Vegas, NV
October 14-17, 2024

Table 1: Clinical estimates

Parameter	Base Case	Range	Distribution	Source
Total population				
Age (mean)	56.3	22 to 81	Normal	Vlaar[2]
Vilobelimab 60-day survival	0.65	0.58 to 0.73	Beta	Vlaar[2]
SoC 60-day survival	0.54	0.48 to 0.62	Beta	Vlaar[2]
Immunomodulator (IM) sub-group population				
Age (mean)	58.6	22 to 79	Normal	Shorr[3]
Vilobelimab 60-day survival	0.82	0.65 to 0.93	Beta	Shorr[3]
SoC 60-day survival	0.51	0.34 to 0.66	Beta	Shorr[3]
Renal replacement therapy (vilobelimab)	0.096	0.086 to 0.106	Beta	Vlaar[2]
Renal replacement therapy (SoC)	0.157	0.141 to 0.173	Beta	Vlaar[2]
Duration of RRT (vilobelimab)	7.8	6.8 to 8.8	Gamma	Vlaar[2]
Duration of RRT (SoC)	9.4	8.1 to 10.7	Gamma	Vlaar[2]

Table 2: Cost estimates

Costs	Base Case	Range	Source
Renal replacement therapy (RRT)	$2,000/day	$1,800 to $2,200	Tseng[7]
Vilobelimab per administration	$6,350	$5,715 to $6,985	RED Book[8]
Intensive care treatment of COVID-19	$100,461	$40,218 to $100,461	Lopez[9]
Premature mortality	$146,494	N/A	Putri[6]

Table 3: Utility and disutility values (Sheinson[5])

	Base case	SD (standard deviation)	Distribution
US age-specific utility			
40-49	0.87	0.002	Beta
50-59	0.84	0.003	Beta
60-69	0.82	0.003	Beta
70-79	0.79	0.004	Beta
>80	0.74	0.006	Beta
Mechanical ventilation during hospitalization disutility	0.56	0.30	Beta
Post-discharge disutility for patients requiring mechanical ventilation			
Year 1	0.130	0.013	Beta
Year 2	0.067	0.007	Beta
Year 3	0.062	0.006	Beta
Year 4	0.026	0.001	Beta

RESULTS

- Total population (see Table 4):
 - Vilobelimab+SoC resulted in 7.99 QALYs vs 6.70 QALYs with SoC alone.
 - Costs were $132,247 for vilobelimab+SoC and $103,414 for SoC alone.
 - ICER for vilobelimab+SoC vs SoC alone was $22,287/QALY.
 - Secondary analysis including cost of premature mortality due to COVID-19 found that ICER was $10,373/ QALY for vilobelimab+SoC vs SoC alone.
- IM subgroup (see Table 4):
 - Vilobelimab+SoC resulted in 9.71 QALYs vs 6.05 QALYs for SoC alone.
 - Costs were $132,247 for vilobelimab+SoC and $103,414 for SoC alone.
 - ICER for vilobelimab+SoC vs SoC alone was $7,892/QALY.

Table 4: Base case analysis

Strategy	Cost	Incremental cost	QALYs	Incremental QALYs	ICER
Total population					
SoC	$103,414	Reference	6.70	Reference	Reference
Vilobelimab	$132,247	$28,833	7.99	1.29	$22,287
Immunomodulators subgroup population					
SoC	$103,414	Reference	6.05	Reference	Reference
Vilobelimab	$132,247	$28,833	9.71	3.65	$7,892

Sensitivity Analysis

One-way Sensitivity Analysis

- The three parameters with the greatest impact on the ICER were:
 - Survival rate for vilobelimab
 - Survival rate for SoC
 - Age

Probabilistic Sensitivity Analysis (PSA)

- CE results for both populations were highly robust to uncertainty:
 - Total population: >81% of draws had a cost per QALY gained below the $50,000 WTP threshold; >92% at $100,000 WTP.
 - IM subgroup: >98% of draws had a cost per QALY gained below the $50,000 WTP threshold.

Figure 2: ICER (total population)

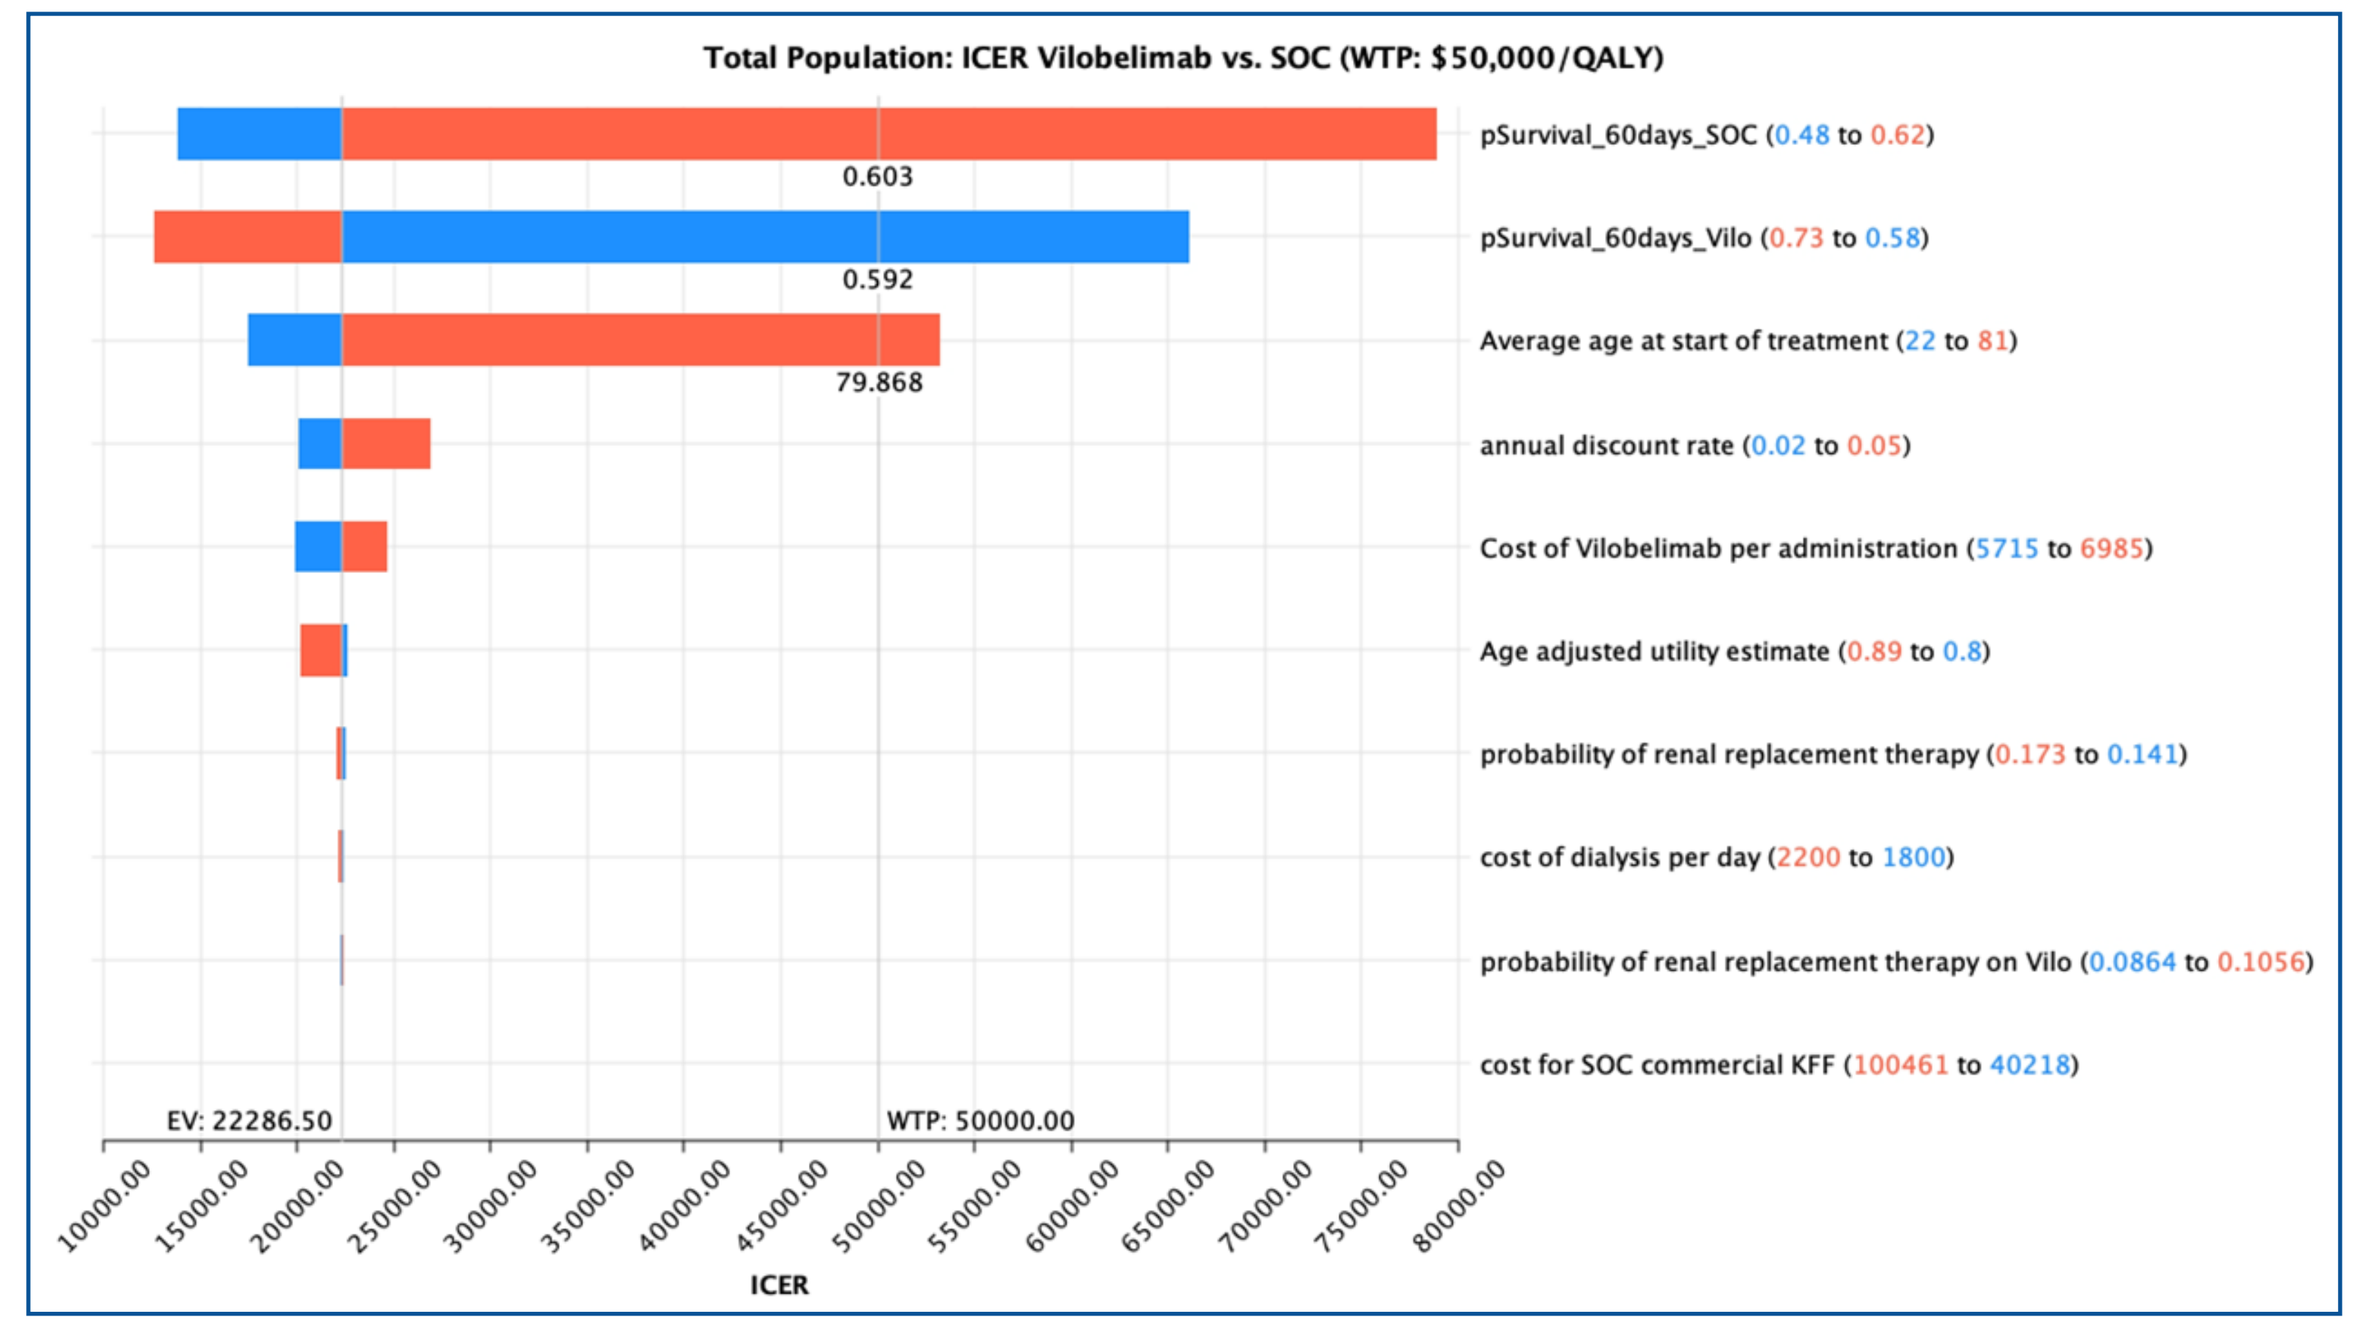
Total Population: ICER Vilobelimab vs. SOC (WTP: $50,000/QALY)
pSurvival_60days_SOC (0.48 to 0.62)
pSurvival_60days_Vilo (0.73 to 0.58)
Average age at start of treatment (22 to 81)
annual discount rate (0.02 to 0.05)
Cost of Vilobelimab per administration (5715 to 6985)
Age adjusted utility estimate (0.89 to 0.8)
probability of renal replacement therapy (0.173 to 0.141)
cost of dialysis per day (2200 to 1800)
probability of renal replacement therapy on Vilo (0.0864 to 0.1056)
cost for SOC commercial KFF (100461 to 40218)
0.603
0.592
79.868
EV: 22286.50
WTP: 50000.00
ICER

Figure 3: ICER (IM subgroup)

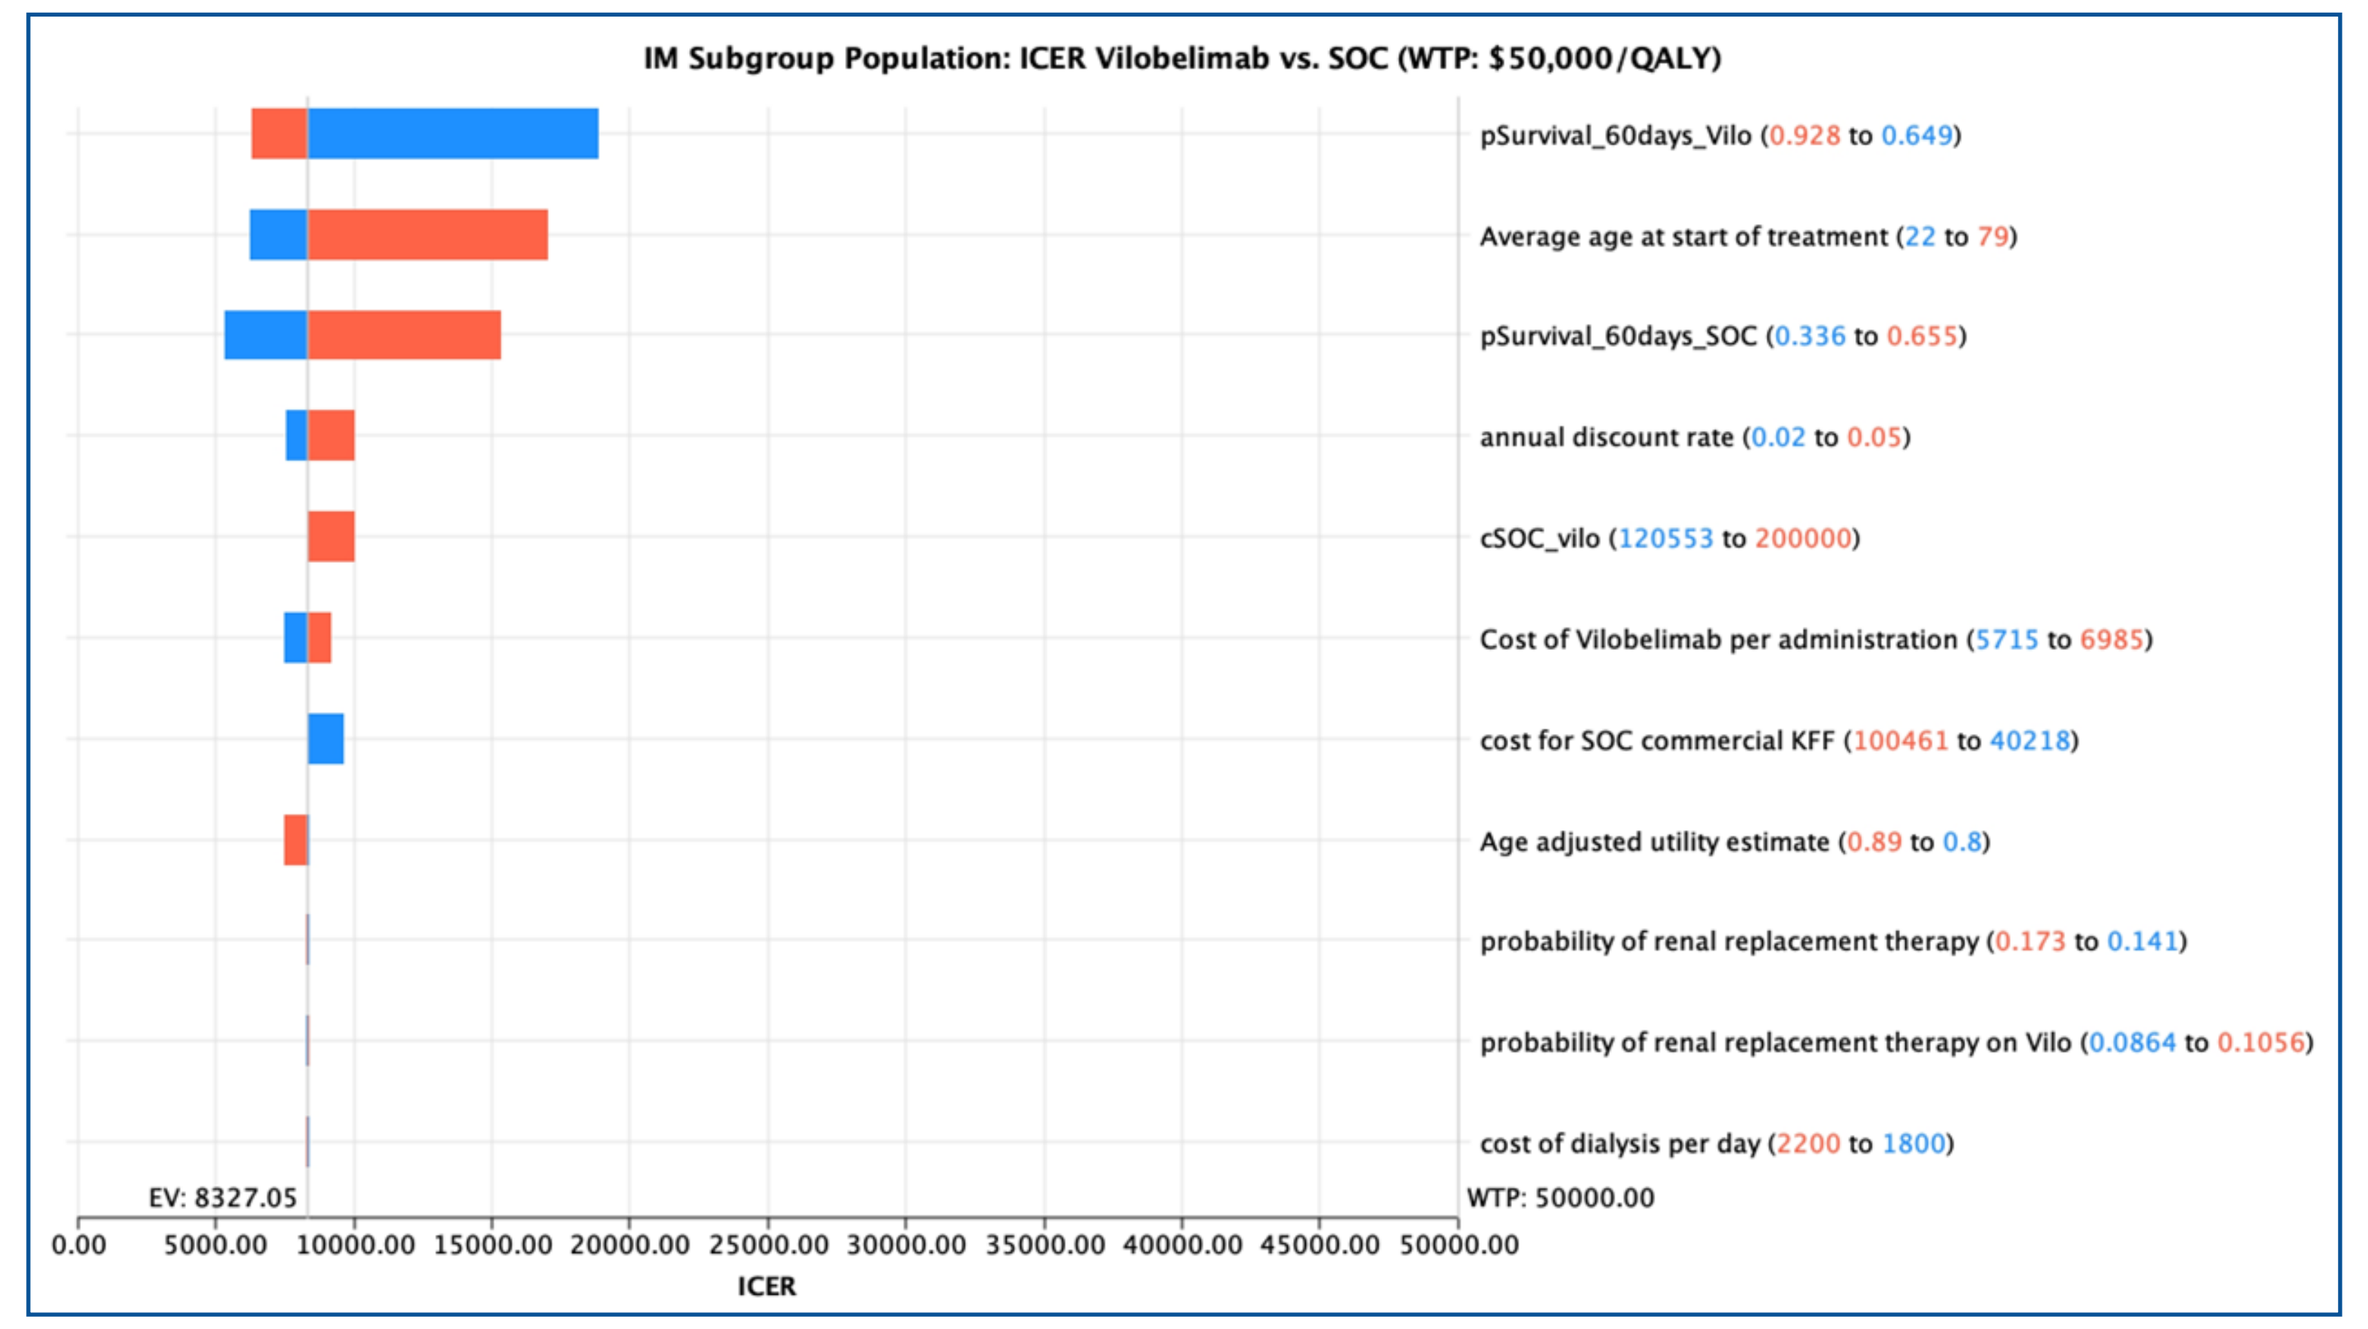
IM Subgroup Population: ICER Vilobelimab vs. SOC (WTP: $50,000/QALY)
pSurvival_60days_Vilo (0.928 to 0.649)
Average age at start of treatment (22 to 79)
pSurvival_60days_SOC (0.336 to 0.655)
annual discount rate (0.02 to 0.05)
cSOC_vilo (120553 to 200000)
Cost of Vilobelimab per administration (5715 to 6985)
cost for SOC commercial KFF (100461 to 40218)
Age adjusted utility estimate (0.89 to 0.8)
probability of renal replacement therapy (0.173 to 0.141)
probability of renal replacement therapy on Vilo (0.0864 to 0.1056)
cost of dialysis per day (2200 to 1800)
EV: 8327.05
WTP: 50000.00
ICER

Figure 4: Incremental Cost-Effectiveness Ratio Scatterplot (Total Population)

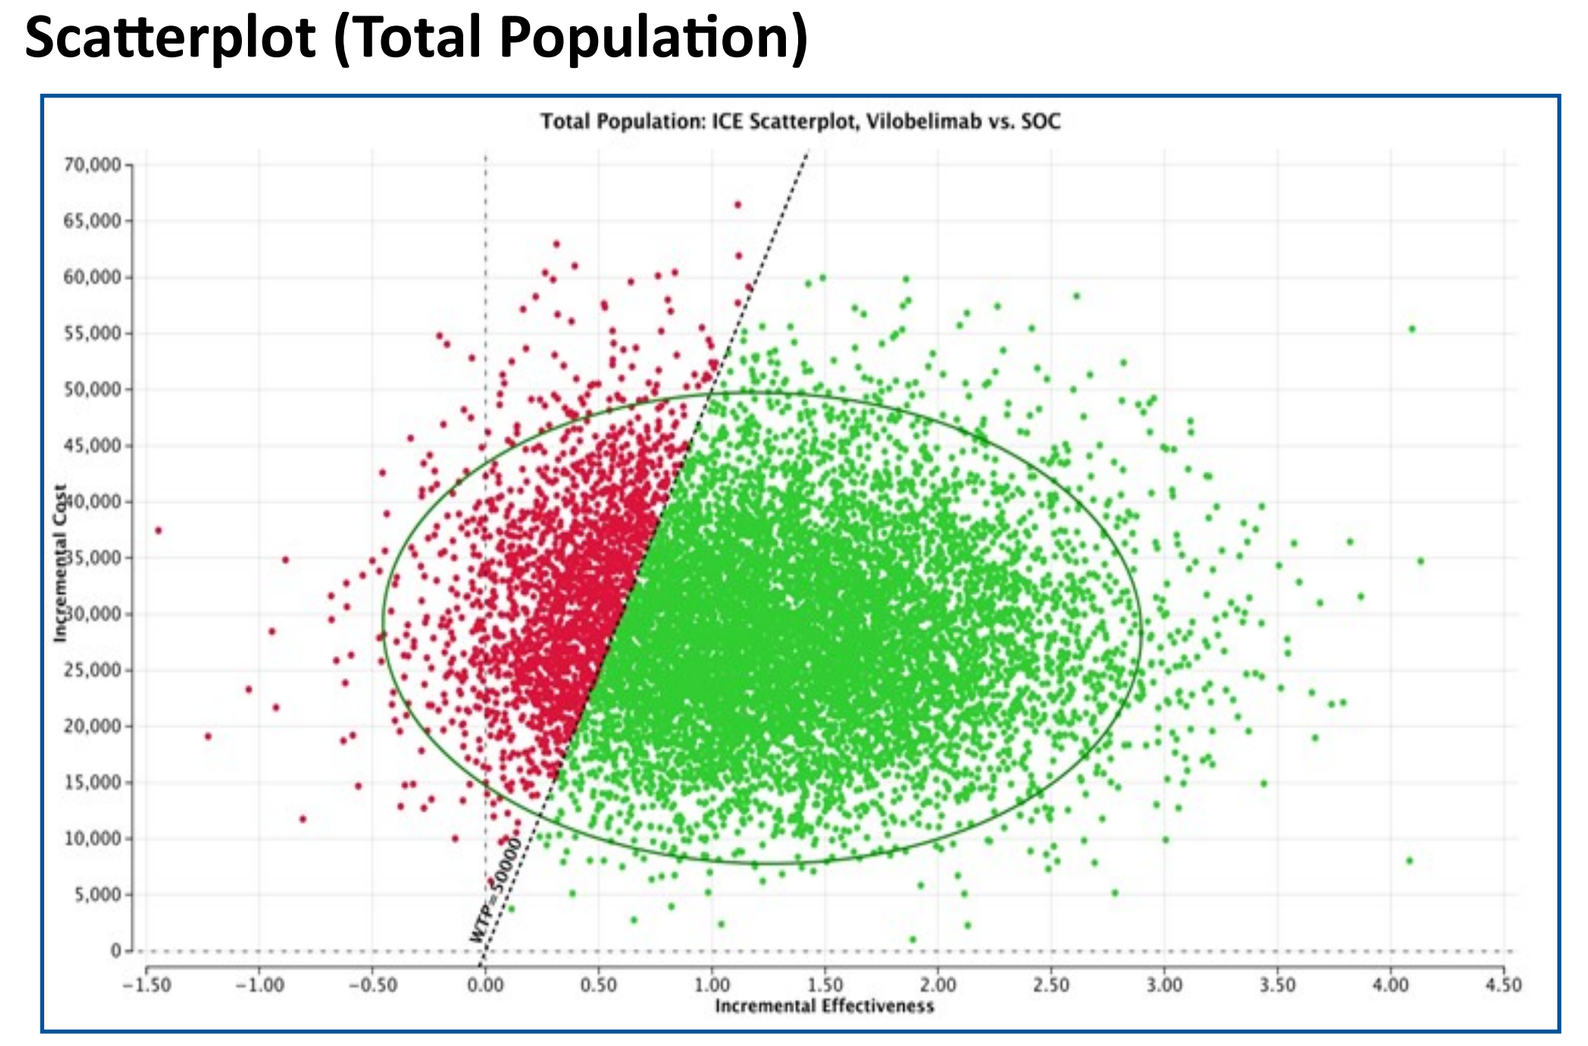
Total Population: ICE Scatterplot, Vilobelimab vs. SOC
Incremental Cost
Incremental Effectiveness

Figure 6: Cost-Effectiveness Ratio Scatterplot (IM Subgroup)

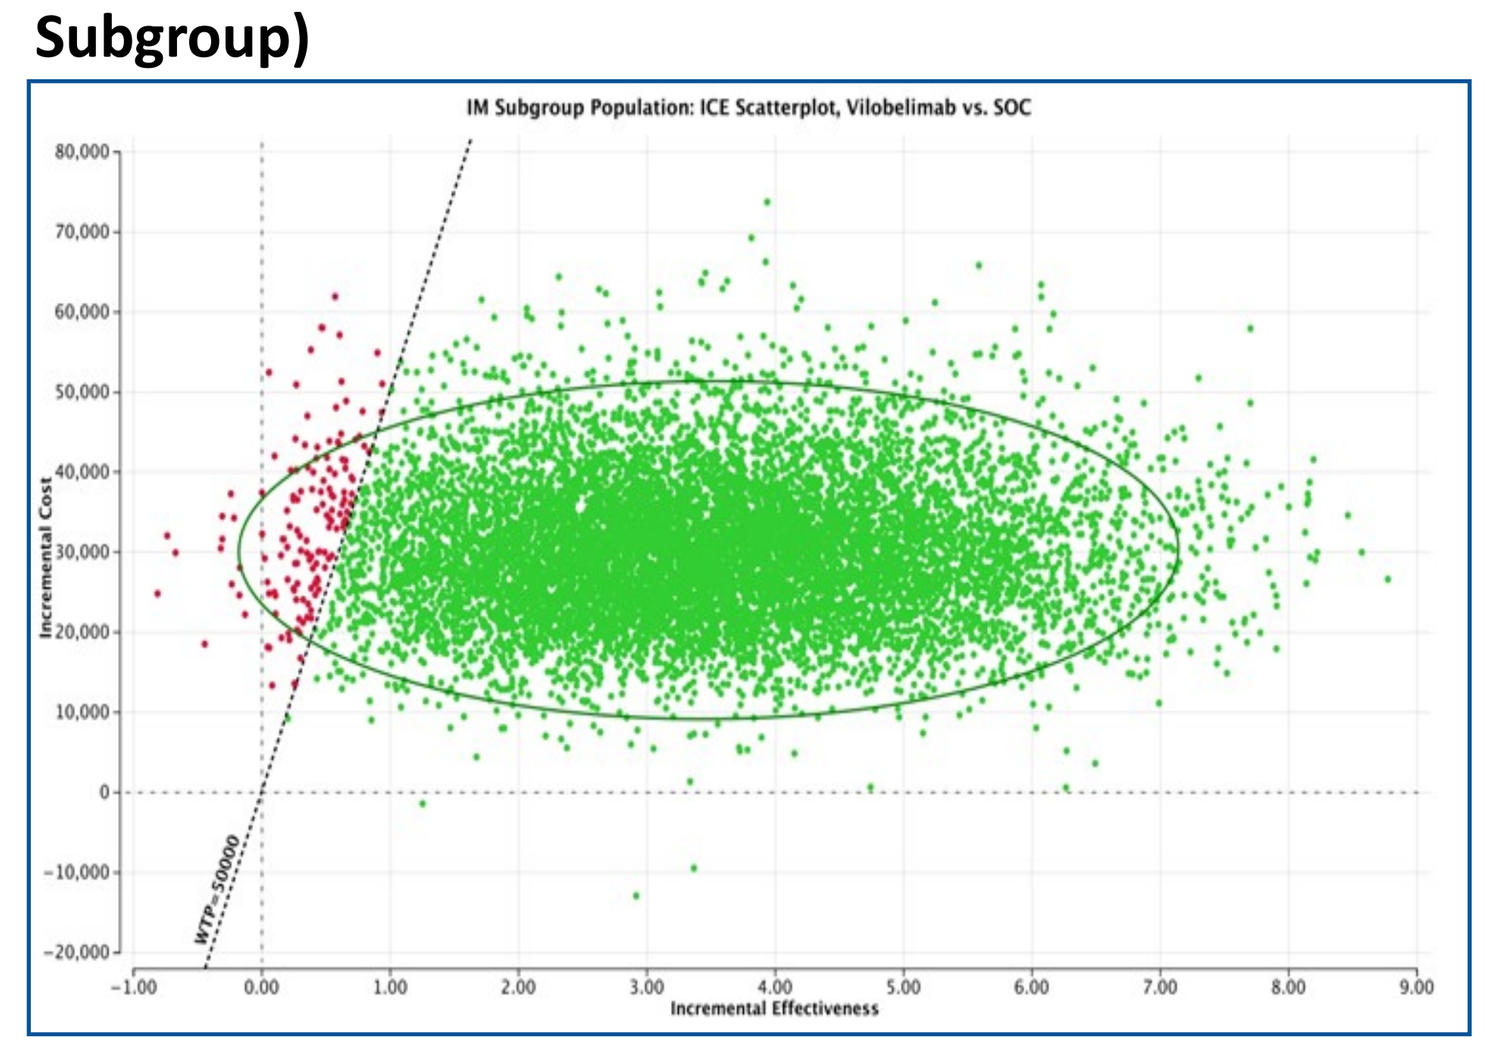
IM Subgroup Population: ICE Scatterplot, Vilobelimab vs. SOC
Incremental Cost
Incremental Effectiveness

Figure 5: Cost-Effectiveness Acceptability Curve (Total Population)

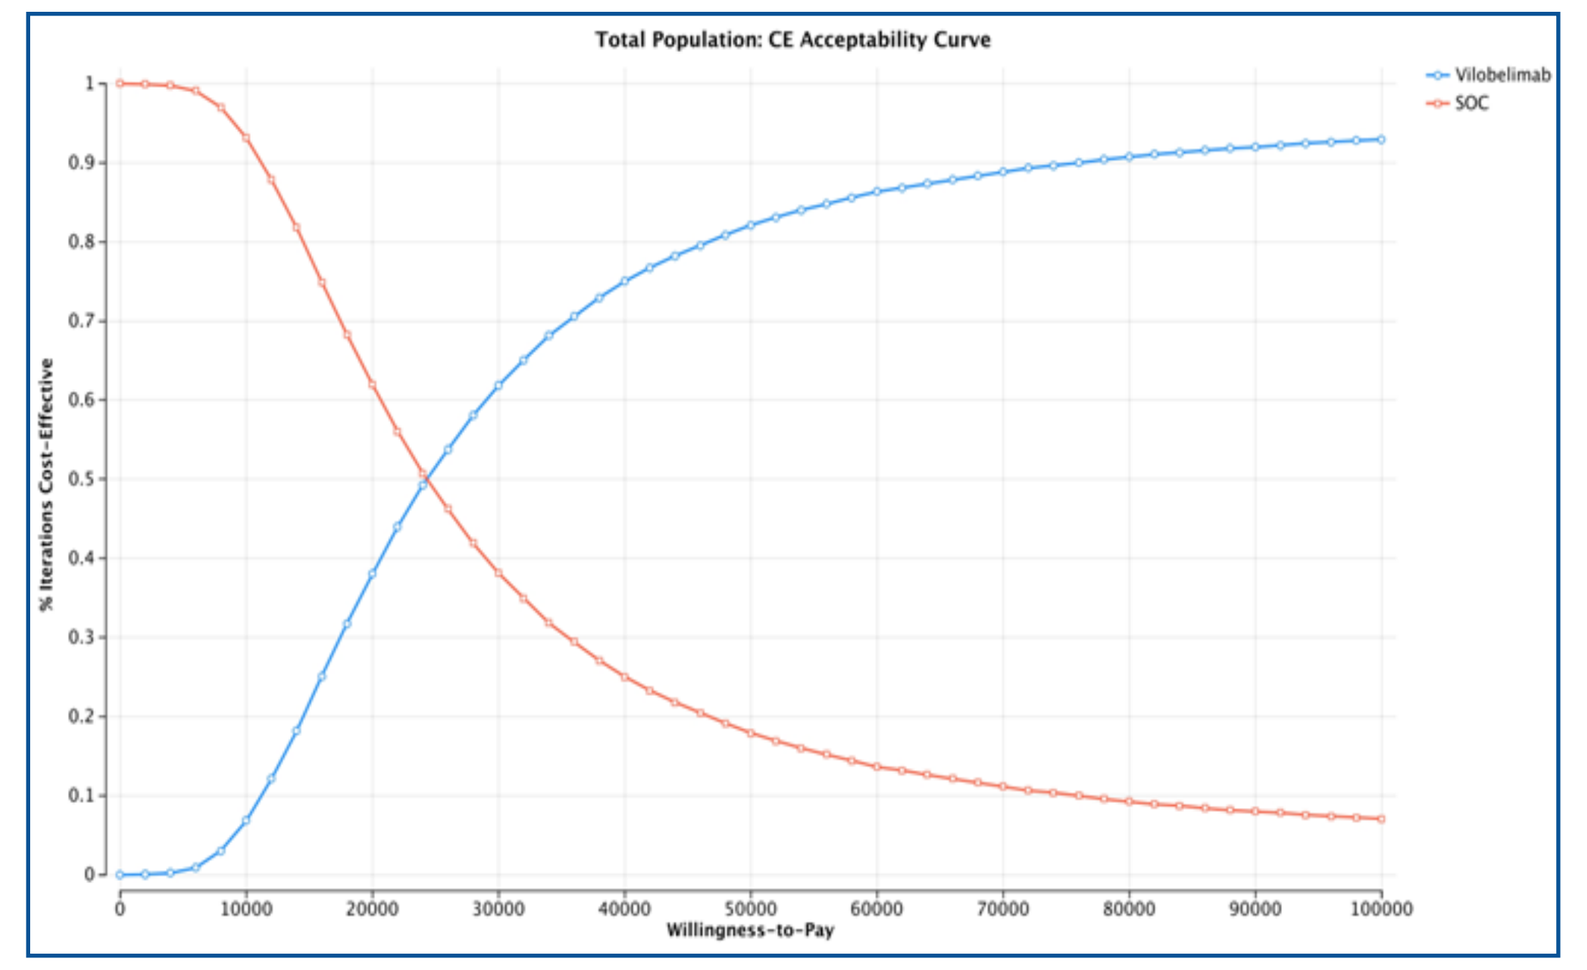
Total Population: CE Acceptability Curve
Vilobelimab
SOC
% Iterations Cost-Effective
Willingness-to-Pay

Figure 6: Cost-Effectiveness Acceptability Curve (IM Subgroup)

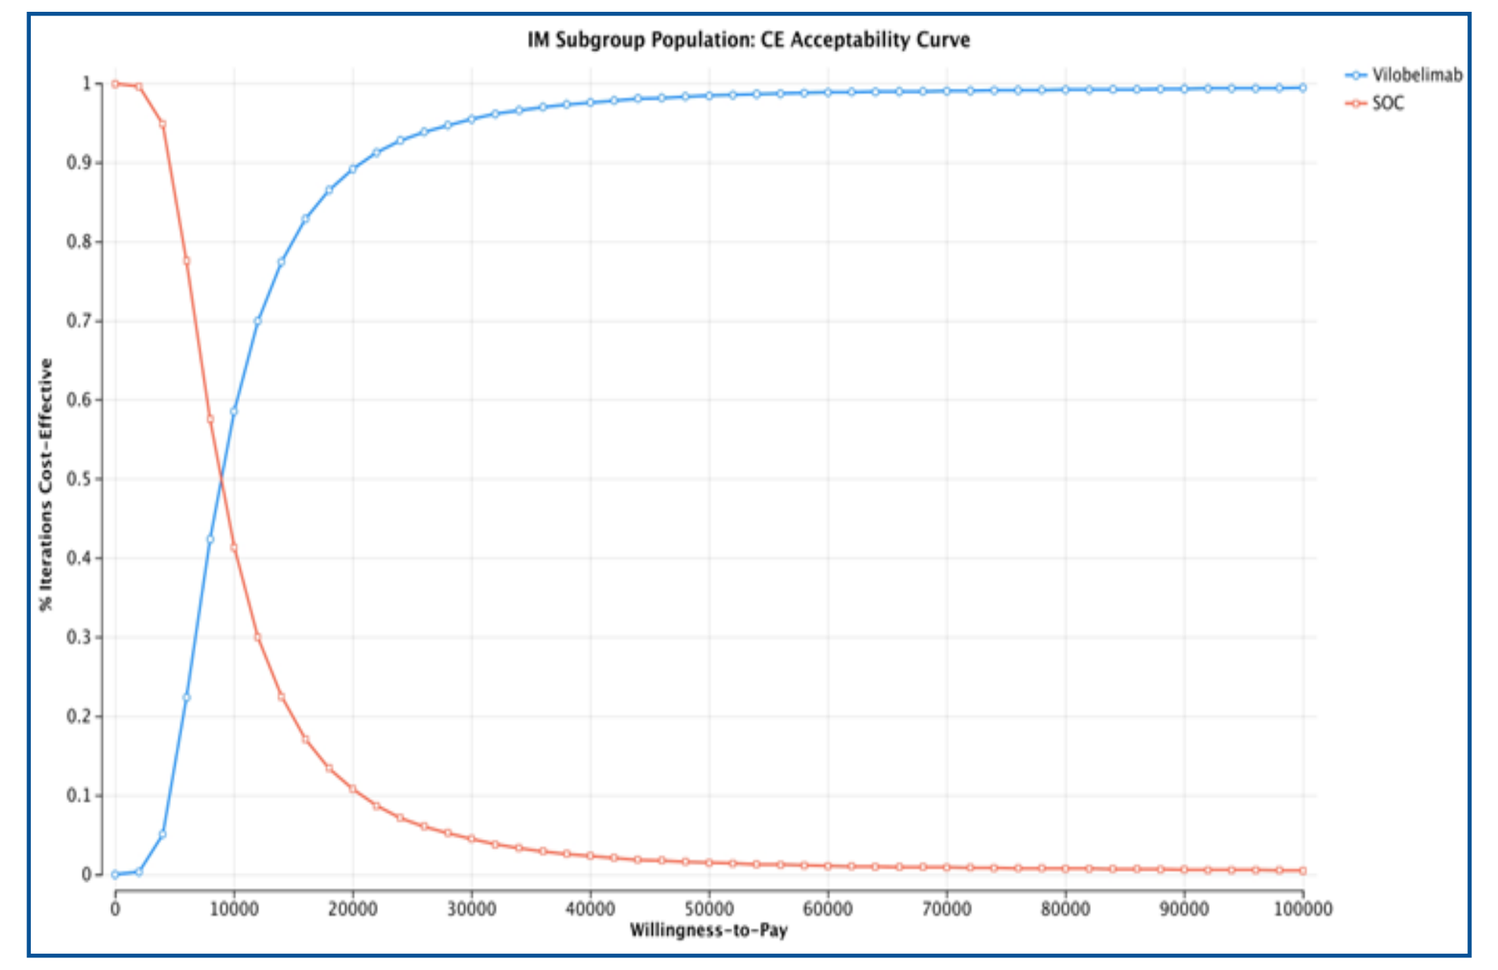
IM Subgroup Population: CE Acceptability Curve
Vilobelimab
SOC
% Iterations Cost-Effective
Willingness-to-Pay

LIMITATIONS

- Model uses a modified societal perspective although data on post-hospital productivity was not included because this information was not available from the study.
- Lifetime perspective was utilized, although mortality data was only available for 28 and 60 days.
- Patients discharged from the hospital were assumed to have a similar life expectancy based on their discharge age, although disutility values were applied for the first 4 years for IMV.
- Hospital costs were assumed to be the same in both groups except for the cost of vilobelimab and RRT.
- Model assumes long-term life expectancy is not affected by receiving IMV due to COVID-19.
- Data used to populate the model was supported by a clinical study conducted outside of the US.

CONCLUSIONS

- Clinical evidence shows vilobelimab has significant mortality benefit in critically ill COVID-19 hospitalized patients requiring IMV.
- Mortality benefit is even greater in patients with prior or concomitant treatment with immunomodulators.
- Using data from the PANAMO Phase 3 study, the cost-effectiveness of vilobelimab add-on therapy to SoC was demonstrated at well below the commonly accepted US willingness-to-pay threshold of $50,000/QALY.

REFERENCES

1. FDA authorizes Gohibic (vilobelimab) injection for the treatment of COVID-19. April 4, 2023. Available at: https://www.fda.gov/drugs/drug-safety-and-availability/fda-authorizes-gohibic-vilobelimab-injection-treatment-covid-19. Accessed May 6, 2023.
2. Vlaar APJ, et al. *Lancet Respir Med*. 2022;10(12):1137-1146.
3. Shorr AF, et al. *Am J Respir Crit Care Med*. 2024;209:A5511.
4. CDC Life Tables. Available at: https://www.cdc.gov/nchs/products/life_tables.htm. Accessed Aug 23,2023.
5. Sheinson D, et al. *Adv Ther.* 2021;38(4):1811-1831.
6. Putri W. *Vaccine*. 2018;36(27):2191-2194.
7. Tseng J. *JAMA Intern Med*. 2018;178(4):572-573.
8. RED BOOK online. Available at: https://www.micromedexsolutions.com Accessed Nov 6, 2023.
9. Lopez E, et al. Available at: https://www.kff.org/medicare/issue-brief/comparing-private-payer-and-medicare-payment-rates-for-select-inpatient-hospital-services/. Accessed Aug 28, 2023.